P.575

Long-term functional improving effects of cariprazine: Post-hoc analyses of acute and predominant negative symptom schizophrenia patient data

István Laszlovszky[1], Károly Acsai[1], Ágota Barabássy[1], Barbara Sebe[1], Balázs Szatmári[1], Willie Earley[2], György Németh[1]

[1]*Gedeon Richter Plc., Budapest, Hungary and* [2]*Abbvie (Allergan), Madison, NJ, USA*

INTRODUCTION

Schizophrenia is a chronic and disabling psychiatric disease characterized by four core symptom domains: positive symptoms, negative symptoms, cognitive impairment and affective symptoms. In addition to symptoms, day-to-day functioning (i.e. personal and social) of patients plays an important role during long-term therapy, to reach a better quality of life.

Improvement of negative symptoms has a substantial impact on function, and treatment of negative symptoms is still a significant unmet medical need.

Cariprazine is a potent dopamine D3 receptor-preferring D2/D3 receptor partial agonist approved by the EMA and FDA for the treatment of schizophrenia, and by the FDA also for manic, depressive and mixed episodes of bipolar I disorder. It proved superior over risperidone in the treatment of predominant negative symptoms (PNS) of schizophrenia.

STUDY OBJECTIVE

To analyze the functional outcome as measured by the Personal and Social Performance (PSP) scale of different patient populations with long-term cariprazine treatment.

METHODS

PSP total scores and subdomains data as secondary/additional outcome measures from two long-term cariprazine treatment studies were post-hoc analyzed using last observation carried forward (LOCF; relapse prevention study) and mixed-effects model for repeated measures (MMRM; PNS study) as per protocol. The p-value of overall treatment effect was not corrected for multiplicity.

In a Phase 3 multinational, randomized, double-blind, placebo-controlled relapse prevention study [1] in adult patients with schizophrenia a total of 765 patients with acute symptoms were treated open-label with cariprazine (3-9 mg/d) for 20 weeks. Stabilized patients were randomized to double-blind placebo (n=99) or cariprazine (n=101) treatment for up to 72 weeks.

In another Phase 3, randomized, double-blind, active-controlled study [2] 456 PNS patients were treated with cariprazine 4.5 mg/d or risperidone 4 mg/d (dose range: 3-6 mg/d for both). The duration of the double-blind treatment time was 26 weeks.

RESULTS

Relapse prevention study (Figure 1 & Table 1)

In the 20 week open-label phase of the relapse prevention study, marked and clinically relevant improvement was seen with cariprazine treatment in those patients who were stabilized and entered into the double-blind phase (19 points PSP total increase). The non-stabilized patients only showed a less than half as much improvement on the PSP total score (8 points increase).

After switching stabilized patients in a double-blind way from cariprazine to placebo, a 7.3 points PSP total score decrease (worsening) occurred in the next 26 weeks and remained stable after it, while in patients continuing cariprazine treatment PSP total score was practically unchanged (0.5 points decrease) for the next 72 weeks.

After switching to placebo, worsening in all PSP subdomains was also present in the next 26 weeks: self-care from 1.46 points at double-blind baseline increased (worsened) by 0.34 points, social relationship from 2.48 by 0.35 points and socially useful activities from 2.70 by 0.37 points.

Figure 1 PSP total score change during cariprazine and placebo treatment

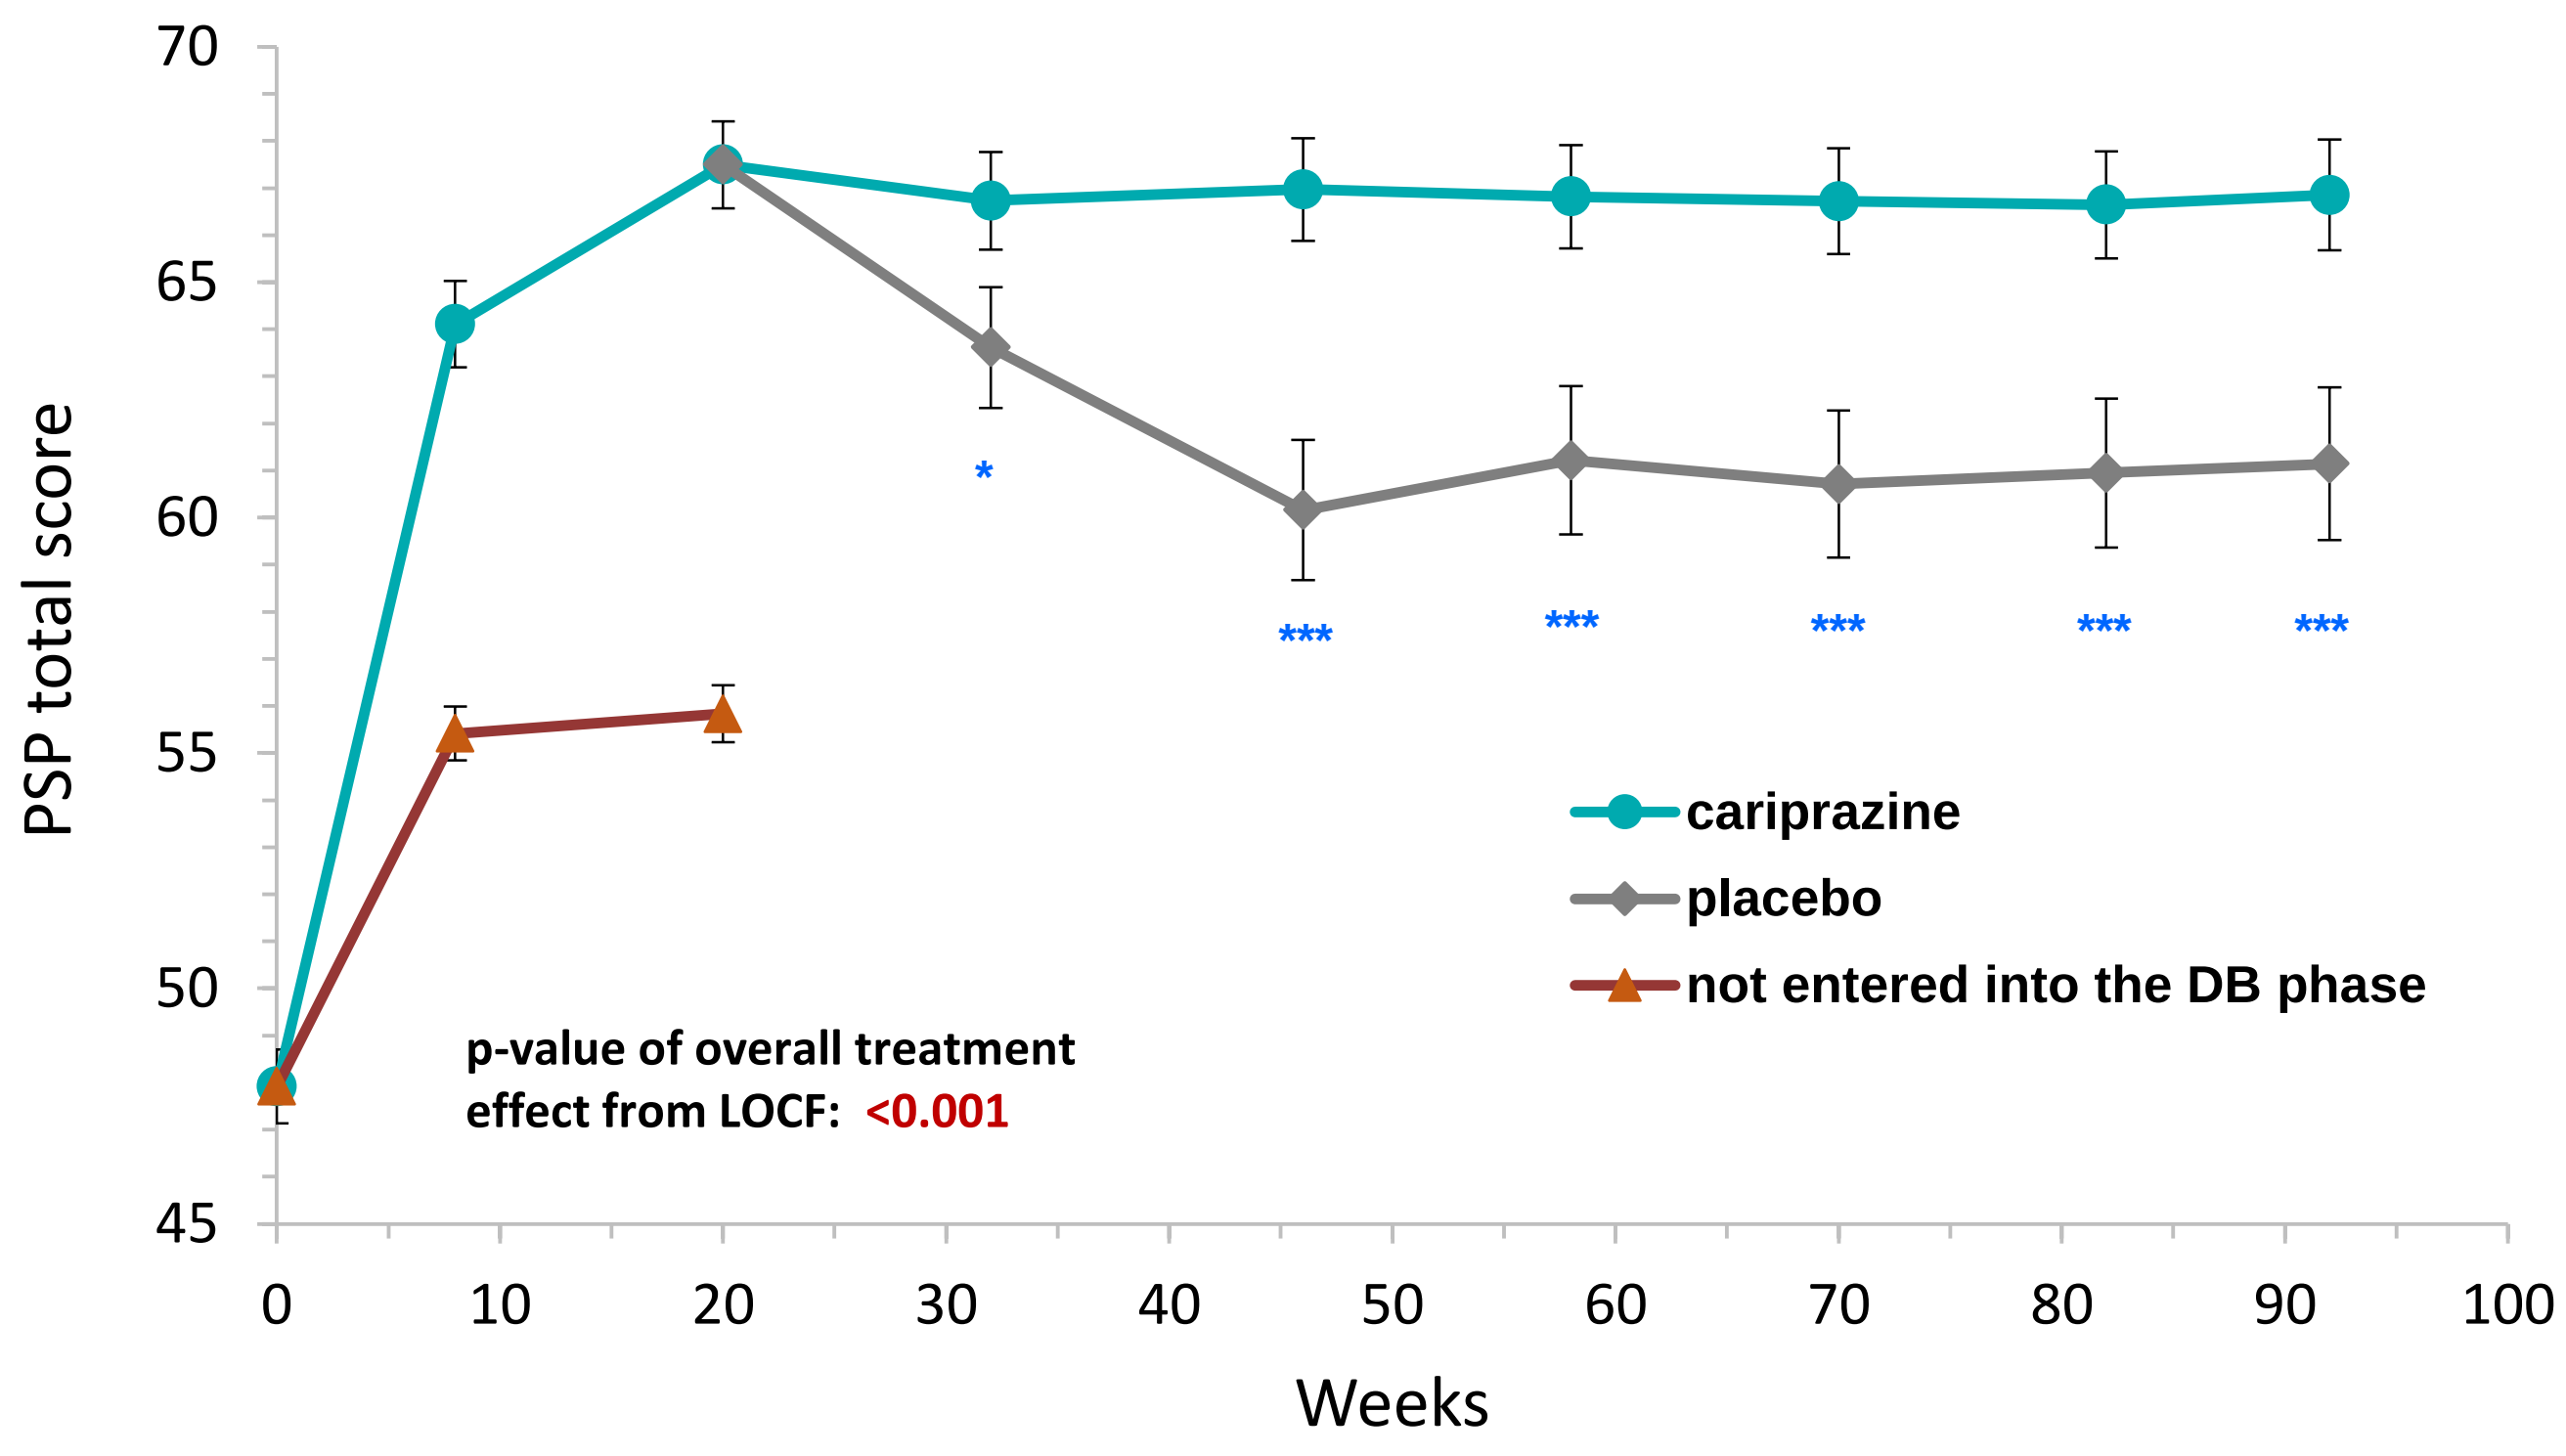
PSP total score
Weeks
cariprazine
placebo
not entered into the DB phase
p-value of overall treatment effect from LOCF: <0.001
*

CONCLUSIONS

- **These post hoc analyses of long-term treatment studies showed that cariprazine is associated with a clinically relevant improvement in patient functioning and social performance compared to either placebo or risperidone.**
- **The results suggest that cariprazine is able to improve not only the positive and negative symptoms of schizophrenia but the every-day functioning and social competence of patients, with the potential to provide a better quality of life in the long run.**

Table 1 Change of PSP sub-domains during different cariprazine treatment

PSP sub-domains Relapse prevention study	Treatment Cariprazine	Placebo	LSMD	P Value
Socially useful activities	-0.03	0.37	0.40	<0.001
Personal and social realtionship	-0.03	0.35	0.38	0.006
Self-care	0.02	0.34	0.32	0.003
Disturbing and aggressive behavior	0.09	0.46	0.38	<0.001
PNS study	**Cariprazine**	**Risperidone**	**LSMD**	**P Value**
Socially useful activities	-0.95	-0.60	-0.35	<0.001
Personal and social realtionship	-0.85	-0.61	-0.24	<0.001
Self-care	-0.70	-0.50	-0.20	0.004
Disturbing and aggressive behavior	-0.06	-0.04	-0.02	0.297

Predominant negative symptom study (Figure 2 & Table 1)

In the PNS study, significantly greater improvements were seen with cariprazine treatment compared to risperidone on the PSP total score (LSMD +4.6) from Week 10 onwards and in the PSP subdomains of self-care, personal and social relationships and socially useful activities (LSMD 0.20, -0.24, 0.35).

Figure 2 PSP total score change during cariprazine and risperidone treatment

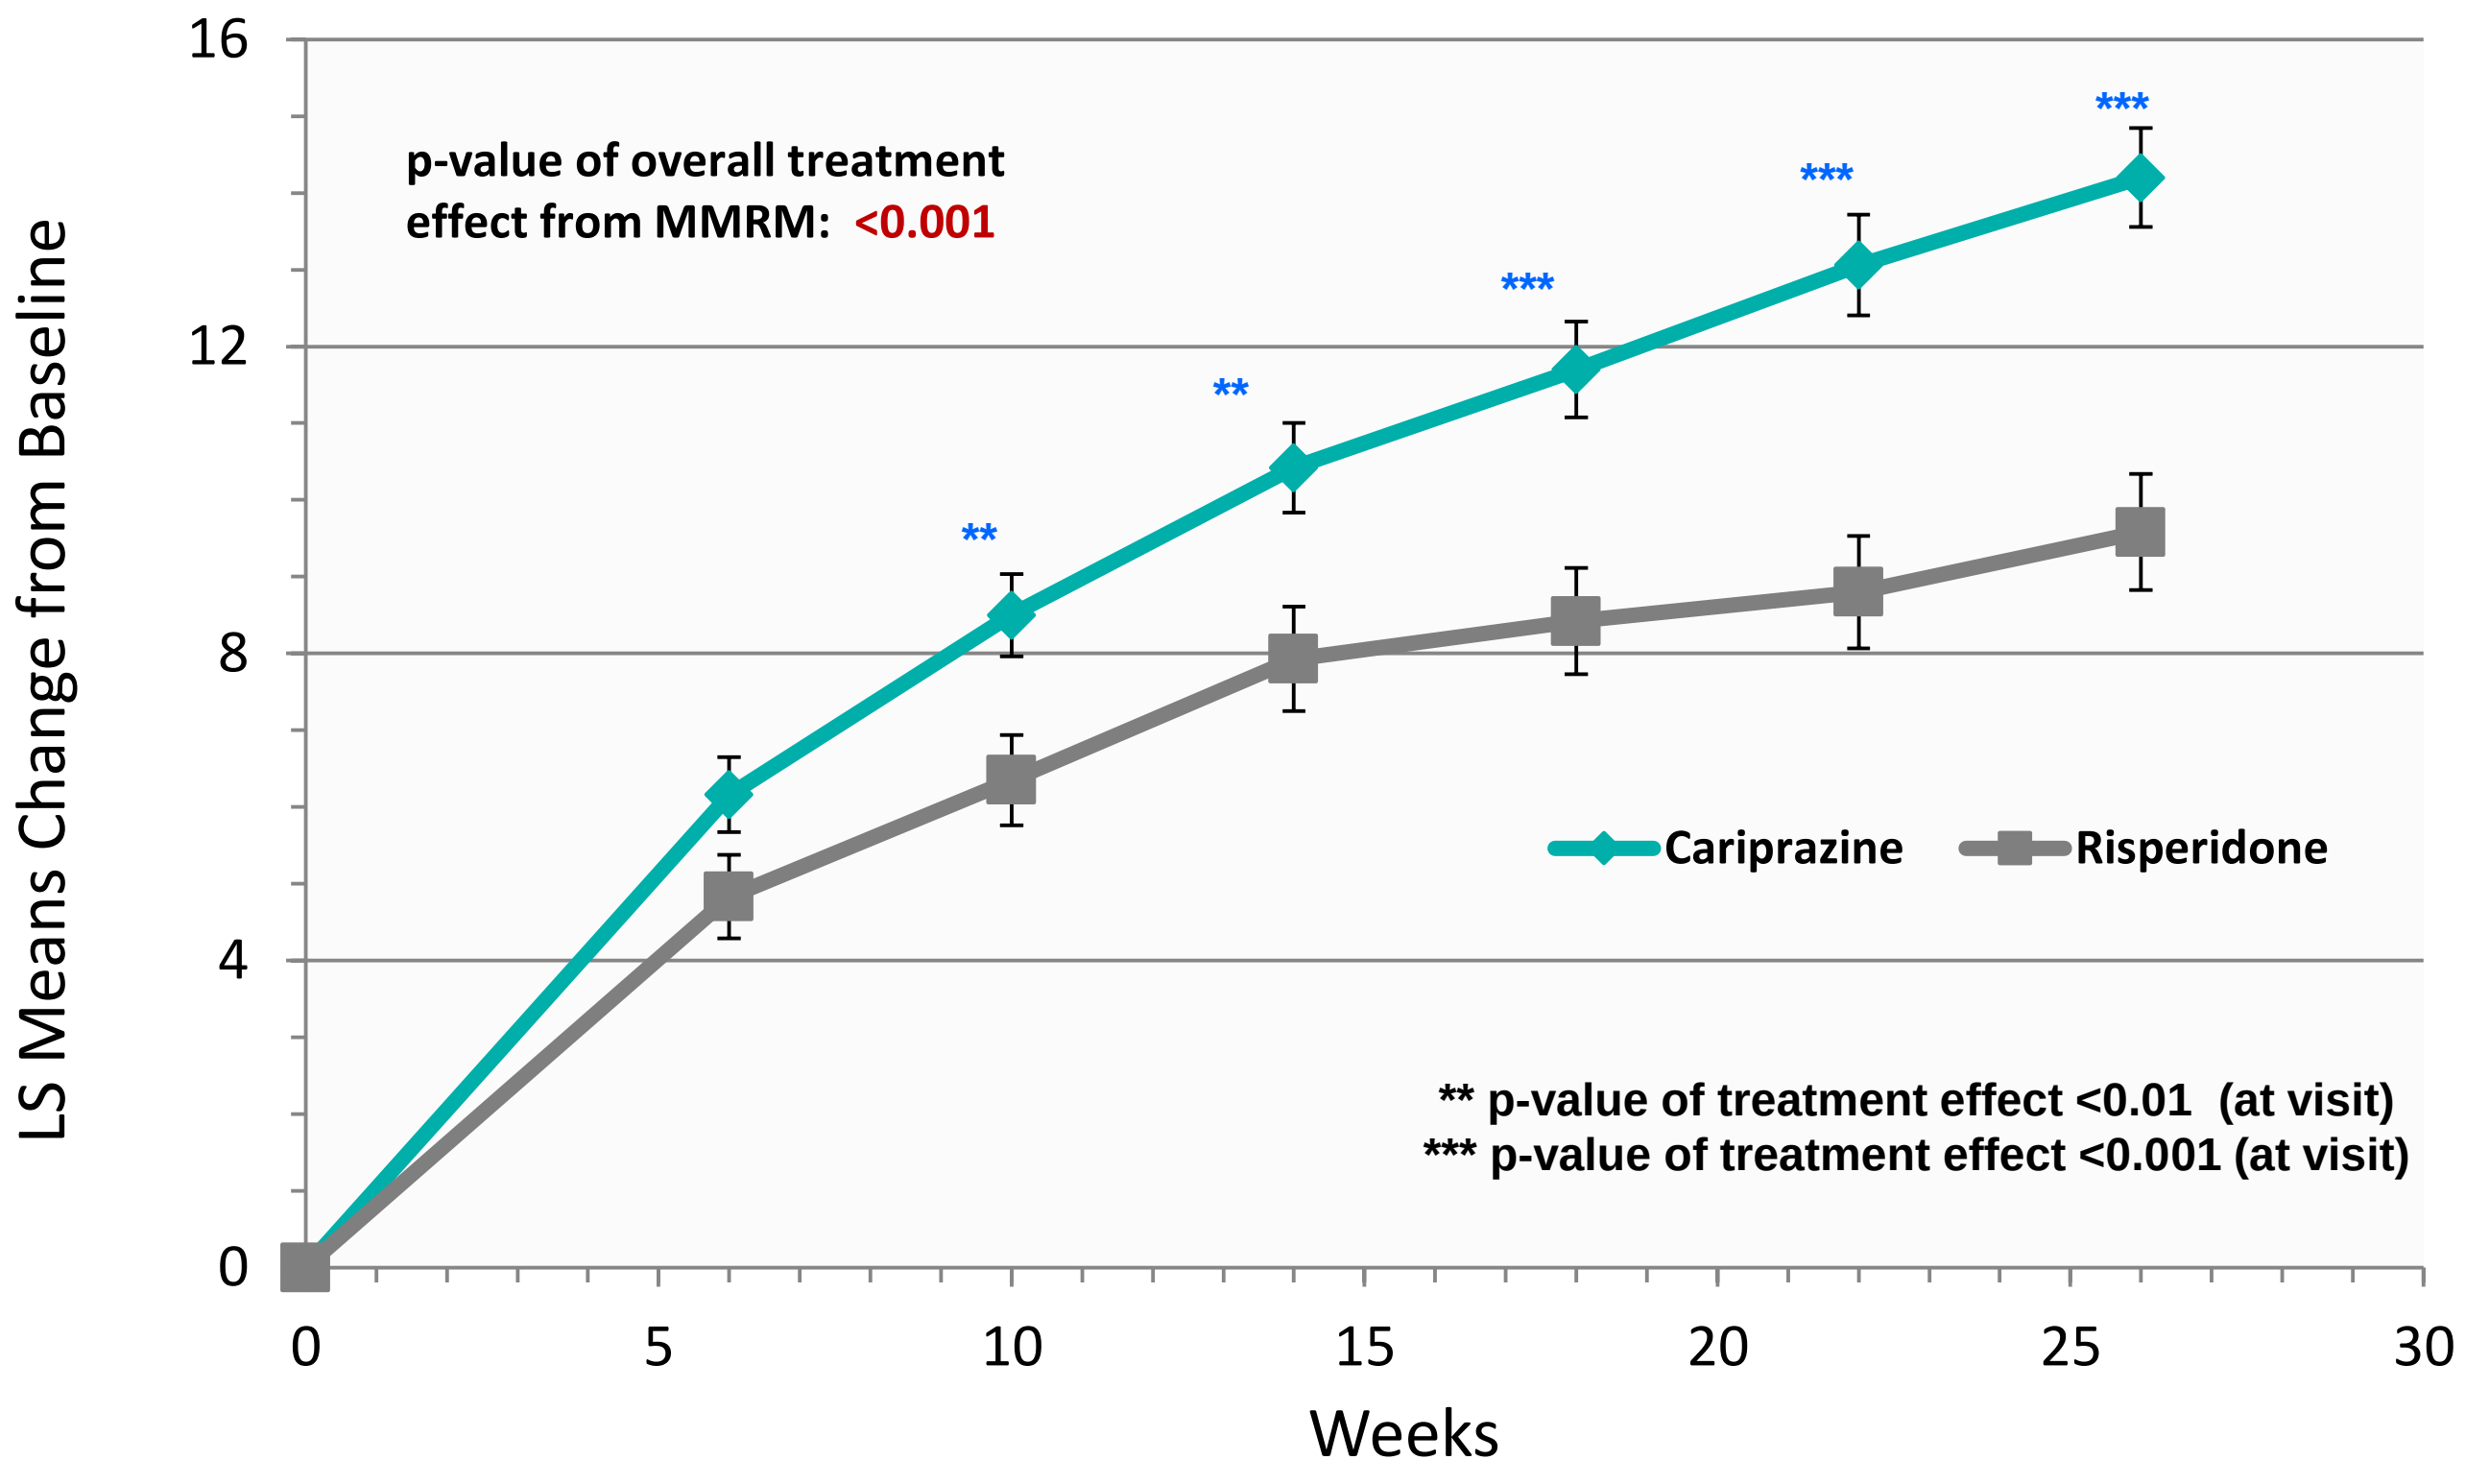
LS Means Change from Baseline
Weeks
p-value of overall treatment effect from MMRM: <0.001
Cariprazine
Risperidone
** p-value of treatment effect <0.01 (at visit)
*** p-value of treatment effect <0.001 (at visit)

REFERENCES

[1] Durgam, S., Earley, W., Li, R., Li, D., Lu, K., Laszlovszky, I., Fleischhacker, W.W., Nasrallah, H.A. Long-term cariprazine treatment for the prevention of relapse in patients with schizophrenia: A randomized, double-blind, placebo-controlled trial. 2016, *Schizophr Res* **176**, 264-271.

[2] Németh, G., Laszlovszky, I., Czobor, P., Szalai, E., Szatmári, B., Harsányi, J., Barabássy, Á., Debelle, M., Durgam, S., Bitter, I., Marder, S., Fleischhacker, W.W. Cariprazine versus risperidone monotherapy for treatment of predominant negative symptoms in patients with schizophrenia: a randomised, double-blind, controlled trial. 2017, *The Lancet* **389**, 1103-1113.

DISCLOSURES & FUNDING STATEMENT

- The studies were funded by Gedeon Richter Plc. and Abbvie (Allergan).
- Dr. Laszlovszky, Mr. Acsai, Dr. Barabássy, Dr. Sebe, Dr. Szatmári and Dr. Németh are employees of Gedeon Richter Plc.
- Dr. Earley is employee of Abbvie (Allergan).

Presented at the 33rd European College of Neuropsychopharmacology, September 12-15, 2020; Virtual