EPP1097

Network analysis of negative symptoms of schizophrenia in acute and in stable patients with persistent, predominantly negative symptoms

K. Demyttenaere,[1] B. Sebe,[2] L. Fonticoli,[3] K. Acsai,[2] A. Barabássy,[2] I. Laszlovszky,[2] B. Szatmári,[2] J. Harsányi,[2] G. Németh[2]

[1]University of Leuven, Faculty of Medicine, Department of Neurosciences, Psychiatry Research Group and University Psychiatric Center KU Leuven, Leuven, Belgium; [2]Richter Gedeon Plc., Medical Division, Budapest, Hungary; [3]Recordati, Medical Affairs Corporate, Milan, Italy

INTRODUCTION

In broad assessments of schizophrenic symptomatology, negative symptoms have repeatedly emerged as a distinct factor with different neurobiological genesis. Additional research focusing on the structure of negative symptoms suggests that this symptom domain seems multidimensional.

Negative symptoms of schizophrenia are critical from two aspects, because

- they are predicting the overall patient functioning [1] and
- they are more difficult to treat [2].

Negative symptoms can be primary or secondary;

- Secondary to primary positive symptoms, depressive symptoms, pharmacological treatment or to hypo stimulation and social deprivation (hospitalization, isolation) [3].

Comparing the network structure of the Positive and Negative Symptom Scale (PANSS) items in populations of patients with and without acute (positive) symptoms could help in differentiating primary from secondary negative symptoms.

STUDY OBJECTIVE

The objective of this post-hoc analysis is to examine the position/connectivity of the of negative symptoms (NS) within the network of the PANSS items in acute as well as in persistent, predominantly negative symptoms (PNS) of schizophrenia.

METHODS

- Analyses are based on two patient groups: acutely ill patients from four phase 2/3 schizophrenia studies [4, 5, 6, 7] and clinically stable patients having persistent predominant negative symptoms with no pseudo-specific factors from a phase 3 clinical trial [8]. Network analyses were done by the baseline scores of each item of PANSS.
- Pooled baseline values were calculated from 4 cariprazine trials in patients with an acute exacerbation of schizophrenia (N=2,193) and a cariprazine/risperidone study in patients with predominantly negative symptoms (N = 461).
- A centrality network analysis was performed on both populations. The R-package “qgraph” [9] was used to compute the partial correlation matrix of the 30 PANSS items.

Figure 1 Network of PANSS symptoms in patients with acute exacerbation of schizophrenia

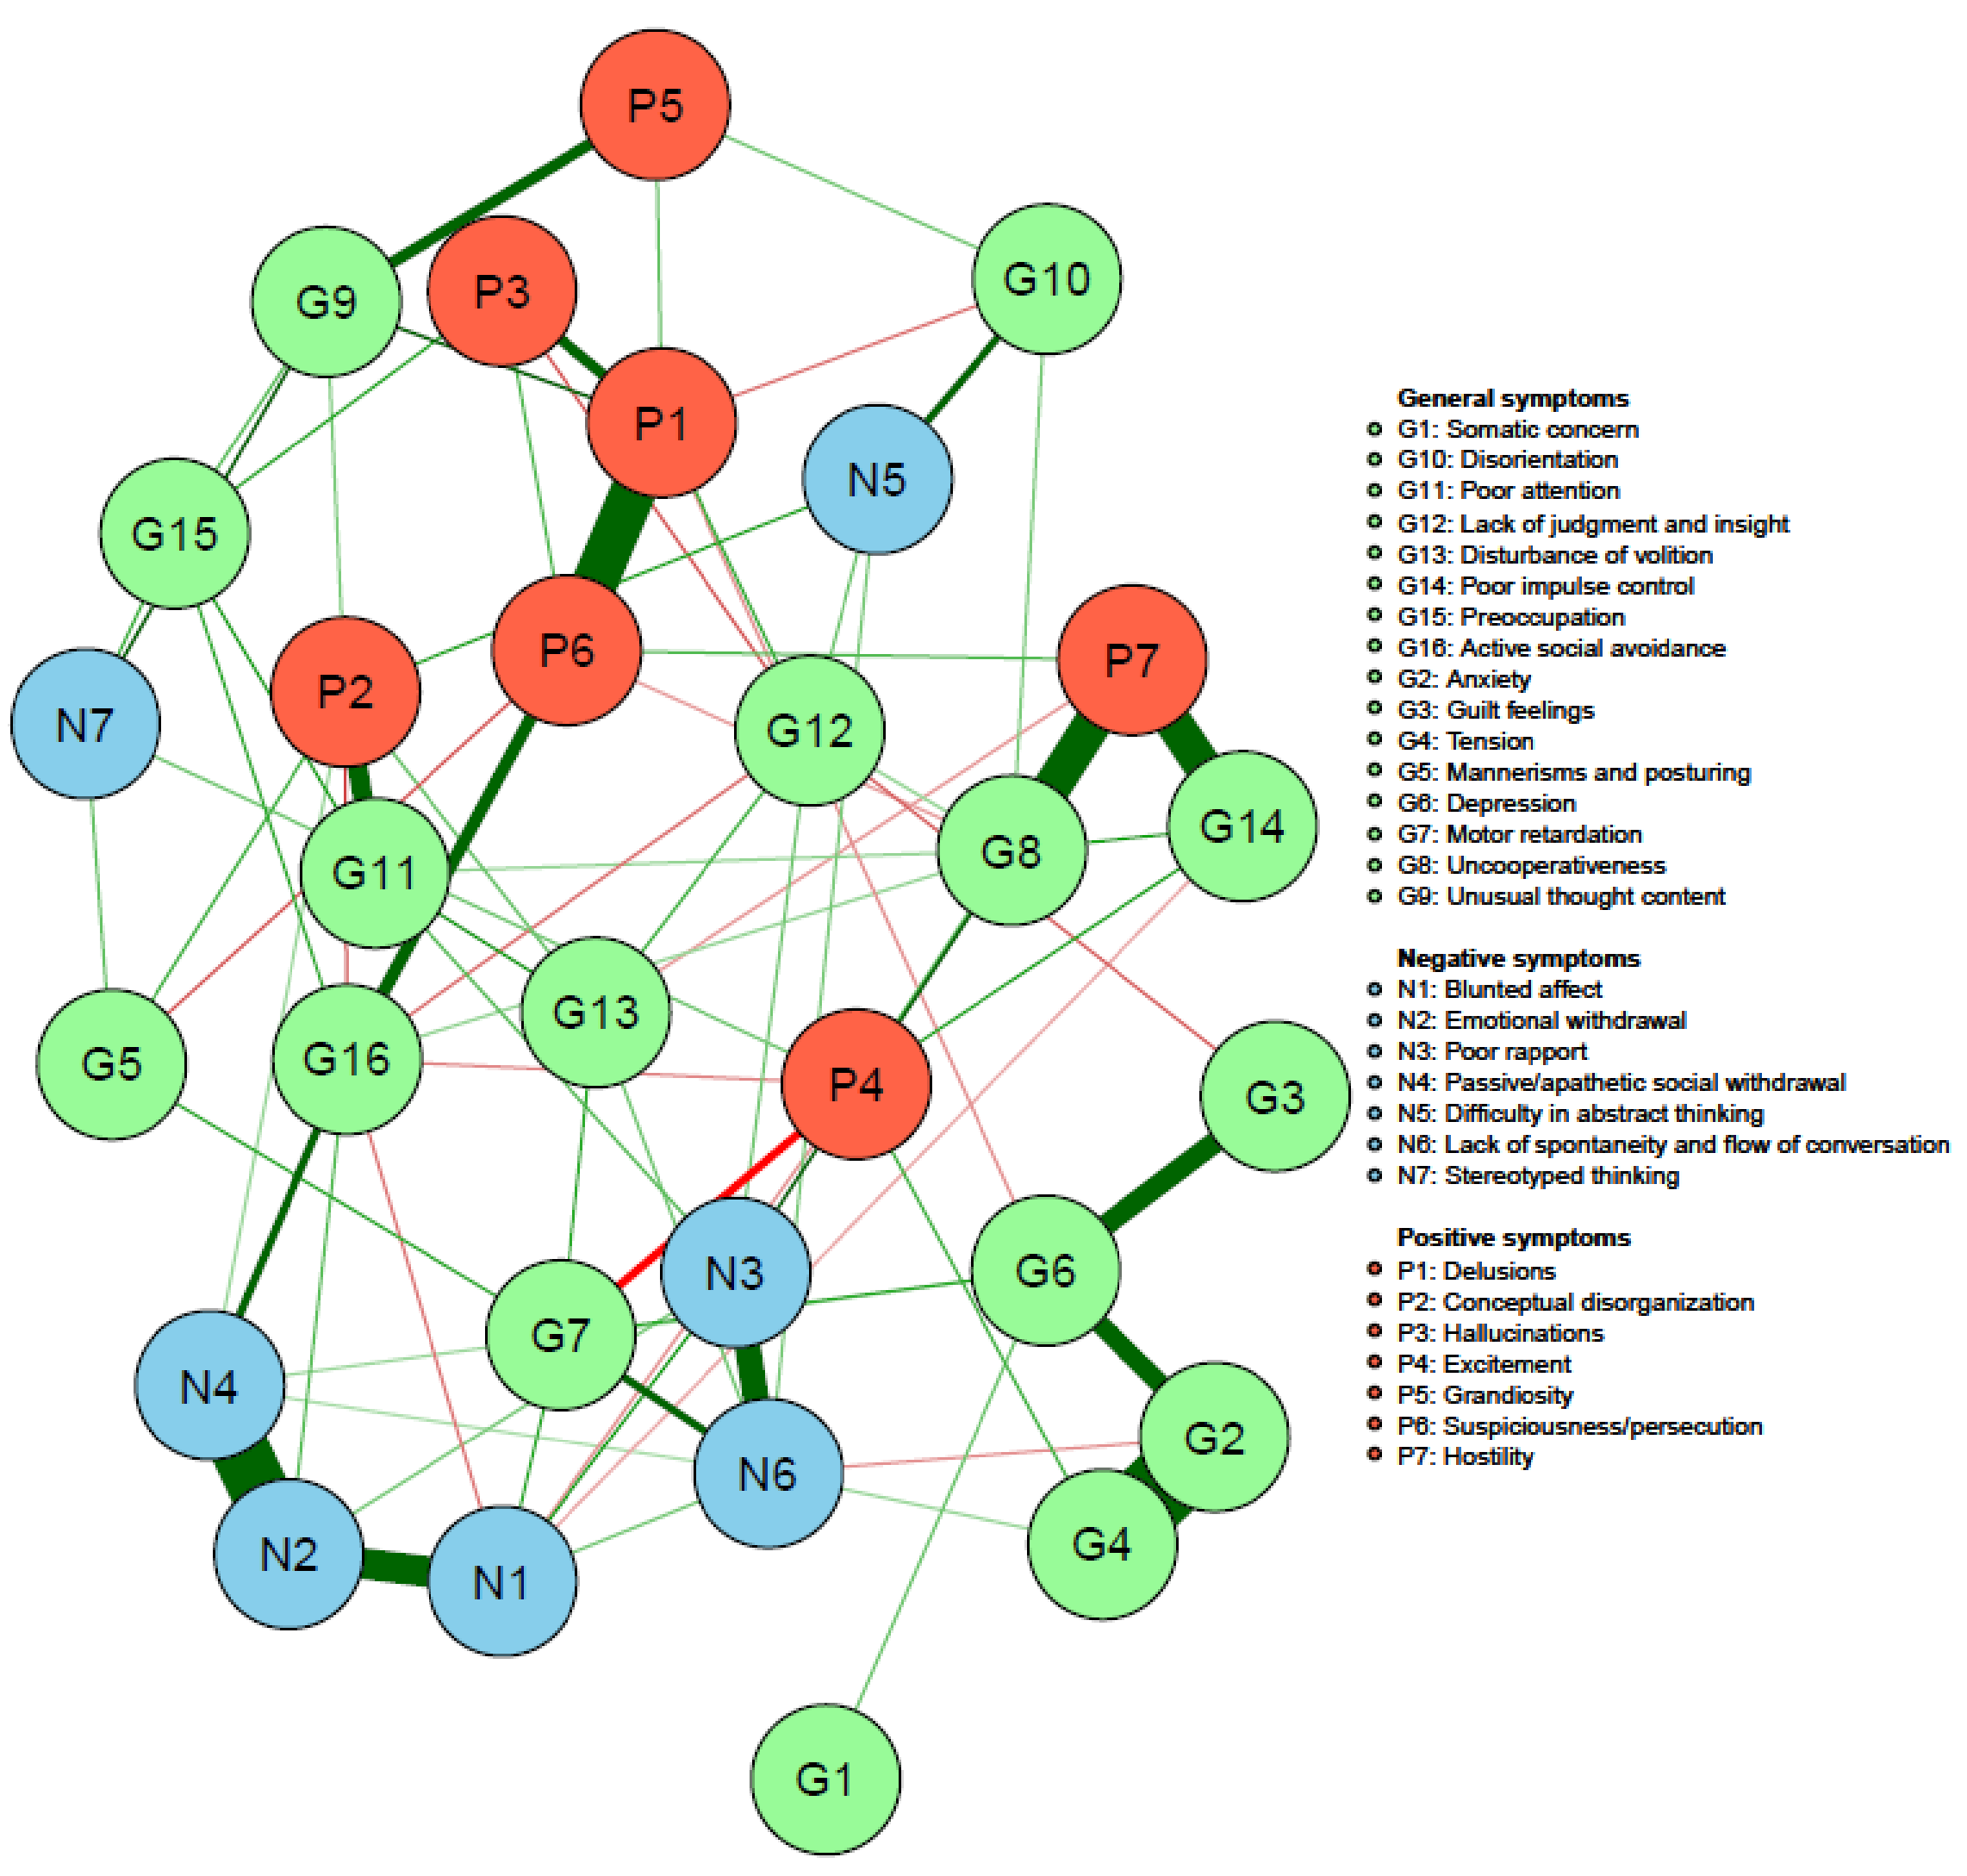
General symptoms
G1: Somatic concern
G10: Disorientation
G11: Poor attention
G12: Lack of judgment and insight
G13: Disturbance of volition
G14: Poor impulse control
G15: Preoccupation
G16: Active social avoidance
G2: Anxiety
G3: Guilt feelings
G4: Tension
G5: Mannerisms and posturing
G6: Depression
G7: Motor retardation
G8: Uncooperativeness
G9: Unusual thought content
Negative symptoms
N1: Blunted affect
N2: Emotional withdrawal
N3: Poor rapport
N4: Passive/apathetic social withdrawal
N5: Difficulty in abstract thinking
N6: Lack of spontaneity and flow of conversation
N7: Stereotyped thinking
Positive symptoms
P1: Delusions
P2: Conceptual disorganization
P3: Hallucinations
P4: Excitement
P5: Grandiosity
P6: Suspiciousness/persecution
P7: Hostility

DISCLOSURES & FUNDING STATEMENT

- Studies were funded by Gedeon Richter Plc. and Allergan Plc.
- Dr. Demyttenaere has received honoraria related to lectures or consultant activities from J&J, LivaNova, Lundbeck, Pfizer, Recordati, and Servier.
- Dr. Sebe, Mr. Acsai, Dr. Barabássy, Dr. Laszlovszky, Dr. Szatmári, Dr. Harsányi and Dr. Németh are employees of Gedeon Richter Plc.
- Dr. Fonticoli is an employee of Recordati.

CONCLUSIONS

- **This network analysis suggests that in patients with an acute exacerbation of schizophrenia negative symptoms are partially primary and partially secondary;**
- **While in patients with predominantly negative symptoms they are mainly primary.**

RESULTS

- The network structure of PANSS symptoms in patients with acute exacerbation of schizophrenia is presented in Figure 1 and in patients with predominantly negative symptoms in Figure 2. The edges are shown by lines connecting the nodes. The width of the lines represents the strength of the association. Green lines represent positive correlations while red lines show negative ones. Nodes with more and stronger connections between each other are located closer to each other, reflecting their relationships in the disease.
- Negative symptom items of the PANSS show different networks in the two groups. In acute patients negative symptom N1 - blunted affect, N2 - emotional withdrawal, N3 - poor rapport, N4 - passive social withdrawal and N6 - lack of spontaneity are well connected to each other and are correlated with several other PANSS symptoms.
- In the PNS population N1, N2, N3, N4 and N6 are completely isolated and are no longer correlated with the other PANSS symptoms in patients with predominantly negative symptoms.
- In both groups two items, N5 - difficulty in abstract thinking and N7 - stereotyped thinking are not connected to NS but are clustered to cognition.

Figure 2 Network of PANSS symptom in patients with predominantly negative symptoms of schizophrenia

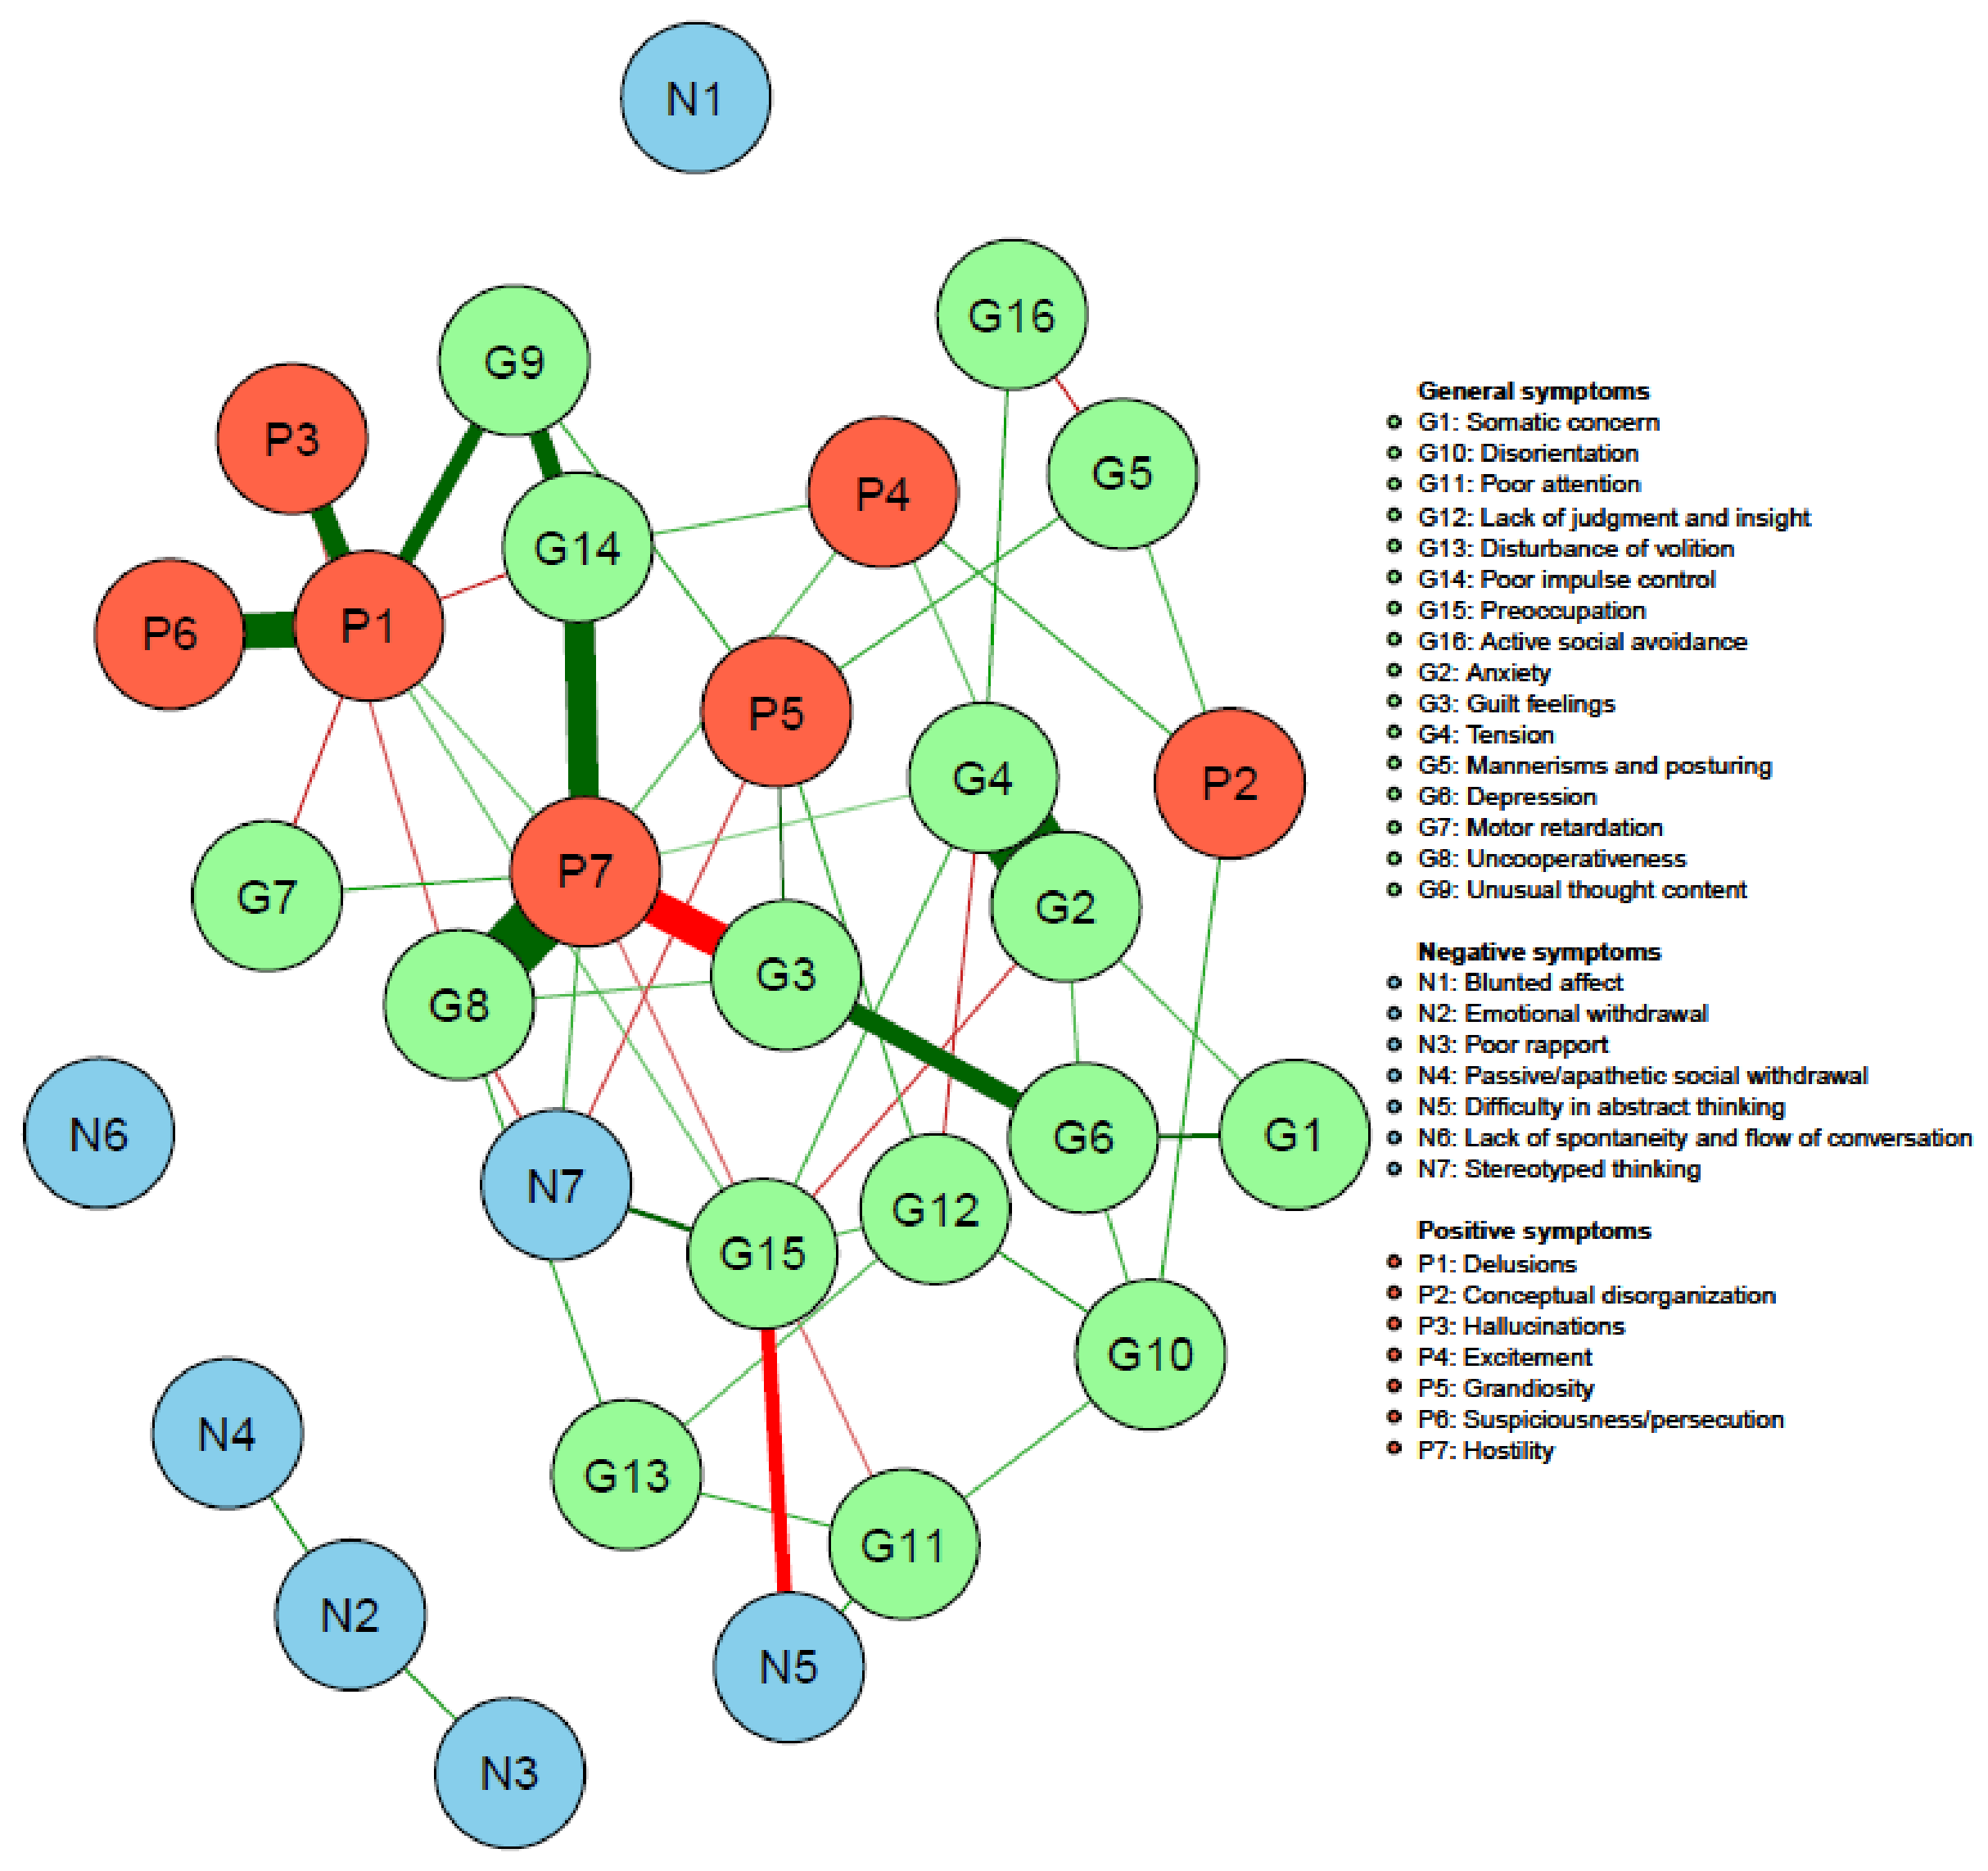
General symptoms
G1: Somatic concern
G10: Disorientation
G11: Poor attention
G12: Lack of judgment and insight
G13: Disturbance of volition
G14: Poor impulse control
G15: Preoccupation
G16: Active social avoidance
G2: Anxiety
G3: Guilt feelings
G4: Tension
G5: Mannerisms and posturing
G6: Depression
G7: Motor retardation
G8: Uncooperativeness
G9: Unusual thought content
Negative symptoms
N1: Blunted affect
N2: Emotional withdrawal
N3: Poor rapport
N4: Passive/apathetic social withdrawal
N5: Difficulty in abstract thinking
N6: Lack of spontaneity and flow of conversation
N7: Stereotyped thinking
Positive symptoms
P1: Delusions
P2: Conceptual disorganization
P3: Hallucinations
P4: Excitement
P5: Grandiosity
P6: Suspiciousness/persecution
P7: Hostility

REFERENCES

[1] Foussias, G. et al., 2014. *Eur Neuropsychopharmacol* **24**, 693–709.
[2] Remington, G. et al., 2016. *Curr Treat Options Psychiatry* **3**, 133–150.
[3] Kirkpatrick, B., 2014. *J Clin Psychiatry* **75**:e09.
[4] Durgam, S. et al., 2016. *Int Clin Psychopharmacol* **31**, 61–68.
[5] Durgam, S. et al., 2014. *Schizophr Res* **152**, 450–457.
[6] Durgam, S. et al. 2015. *J Clin Psychiatry* **76**, e1574-1582.
[7] Kane, J.M. et al., 2015. *J Clin Psychopharmacol* **35**, 367–373.
[8] Németh, G., et al., 2017. *Lancet* **389**, 1103-1113.
[9] Epskamp, S. et al., 2018. *Behav Res* **50**:195–212.

Presented at the 28th European Congress of Psychiatry 2020